Rational Design of ACR-2316, a Novel, Potent WEE1/PKMYT1 Inhibitor with Superior Single Agent Activity Using Acrivon Predictive Precision Proteomics (AP3)

Acrivon Therapeutics

Joelle Baddour-Sousounis[1], Martina Pasetto[2], Helén Nilsson[2], Lei Shi[1], Subodh Kumar[1], Portia Lombardo[1], Zachary Best[1], Kate Rappard[1], Calvin Yang[1], Reina Improgo[1], Everett Hay[1], Uthira Muralitharan[2], Nina Lipjankić[2], Ruban Cornelius[2], Ignacio Arribas Diez[2], Georgia Mitsa[2], Mohamed Eldeeb[2], Valentina Siino[2], Ayesha Murshid[1], Michail Shipitsin[1], Caroline Wigerup[2], Magnus E. Jakobsson[2], Joon Jung[1], David A. Proia[1], Kristina Masson[2], Peter Blume-Jensen[1]

[1]Acrivon Therapeutics Inc., Watertown, MA, USA , [2]Acrivon AB, Medicon Village, Lund, Sweden

Abstract # 330

Board # PB318

Abstract

Coordinated activation of the kinases CDK1, CDK2, and PLK1 is essential for cell cycle progression, while WEE1 and PKMYT1 negatively regulate CDK1/2 through inhibitory phosphorylation. Cancer cells are frequently G1/S checkpoint deficient rendering them vulnerable to WEE1 and PKMYT1 inhibition. Acrivon Predictive Precision Proteomics (AP3) platform is engineered to measure compound-specific effects on the entire tumor cell protein signaling network and drug-induced resistance mechanisms in an unbiased manner in the intact cellular setting. These distinctive capabilities enable AP3's direct application for drug design optimization for monotherapy activity. Using our AP3 platform, we uncovered dominant WEE1 inhibitor-induced resistance mechanisms and demonstrated that these could be quenched by PKMYT1 inhibition. These data support the rationale for a balanced, dual WEE1/PKMYT1 inhibition to achieve potent anticancer activity. Here, we report the in-depth characterization of ACR-2316, a novel, potentially best-in-class dual WEE1/PKMYT1 inhibitor rationally designed for superior single-agent activity and selectivity uniquely enabled by AP3. Methods: Activation of CDK1, CDK2 and PLK1 was assessed by AP3 mass spectrometry-based proteomic profiling through consensus substrate motif analyses and validated using cellular kinase assays. Effects on replication stress, DNA damage and apoptotic cell death were assessed by flow cytometry, cytotoxicity assays, and immunohistochemistry in ovarian cancer cells and xenografts. In vivo efficacy was evaluated in cell lines and patient derived xenograft (PDX) tumors.

Acrivon Predictive Precision Proteomics Reveals Actionable WEE1 Inhibitor-induced Resistance Mechanisms

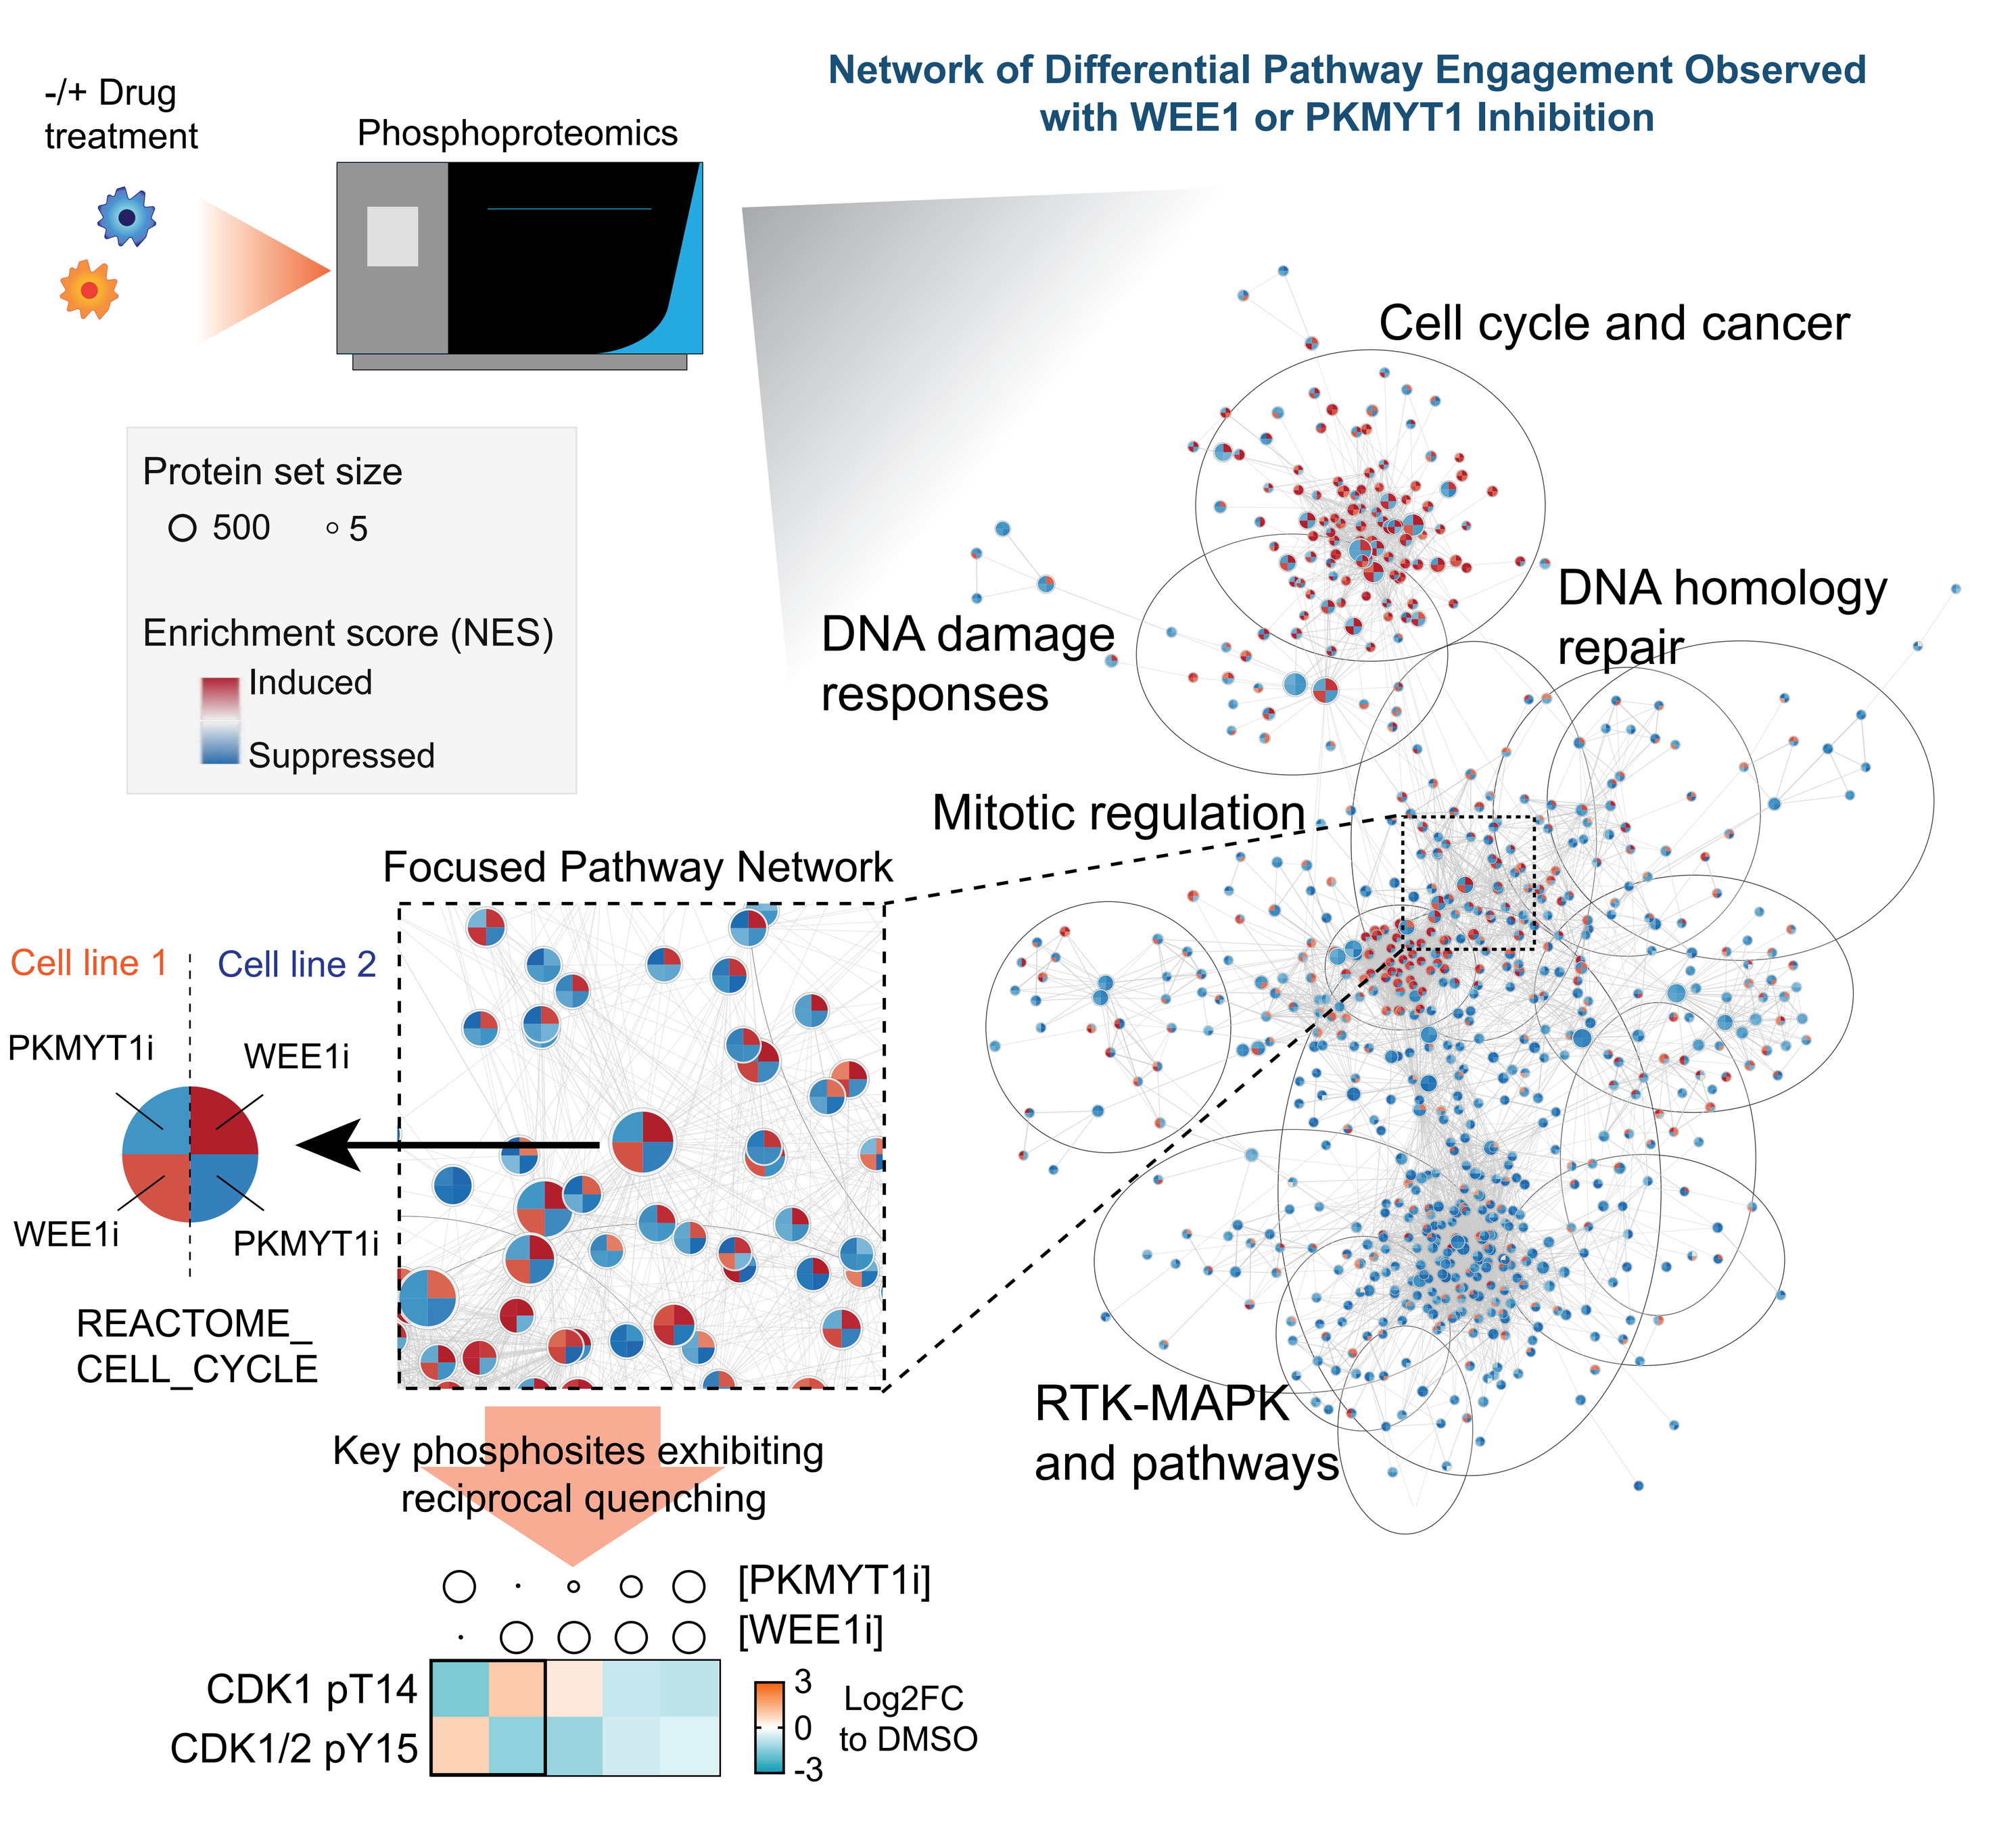
-/+ Drug treatment
Phosphoproteomics
Network of Differential Pathway Engagement Observed with WEE1 or PKMYT1 Inhibition
Protein set size
500
5
Enrichment score (NES)
Induced
Suppressed
Cell cycle and cancer
DNA damage responses
DNA homology repair
Mitotic regulation
RTK-MAPK and pathways
Focused Pathway Network
Cell line 1
Cell line 2
PKMYT1i
WEE1i
WEE1i
PKMYT1i
REACTOME_CELL_CYCLE
Key phosphosites exhibiting reciprocal quenching
[PKMYT1i]
[WEE1i]
CDK1 pT14
CDK1/2 pY15
Log2FC to DMSO

Human tumor cell lines were treated with Adavosertib (200 nM) or Lunresertib (20 nM) for 60 min. Cell lysates were processed for phosphoproteomics mass spectrometry. The drug regulated phosphoproteome was analysed and mapped to cellular pathways to identify resistance mechanisms to WEE1 inhibition. Pathways enriched by WEE1 inhibition included those related to cell cycle, DNA damage response, and mitotic regulation. A subset of these pathways were quenched by PKMYT1 inhibition, with a key phosphosite, CDK1 T14, exhibiting such reciprocal quenching. These data suggest that a combination of WEE1 and PKMYT1 inhibition would be synergistic in terms of fully activating CDK1 and overcoming resistance to WEE1 inhibition.

ACR-2316 Results in Stalled DNA Replication, Induction of DNA Damage and Premature Mitotic Cell Death

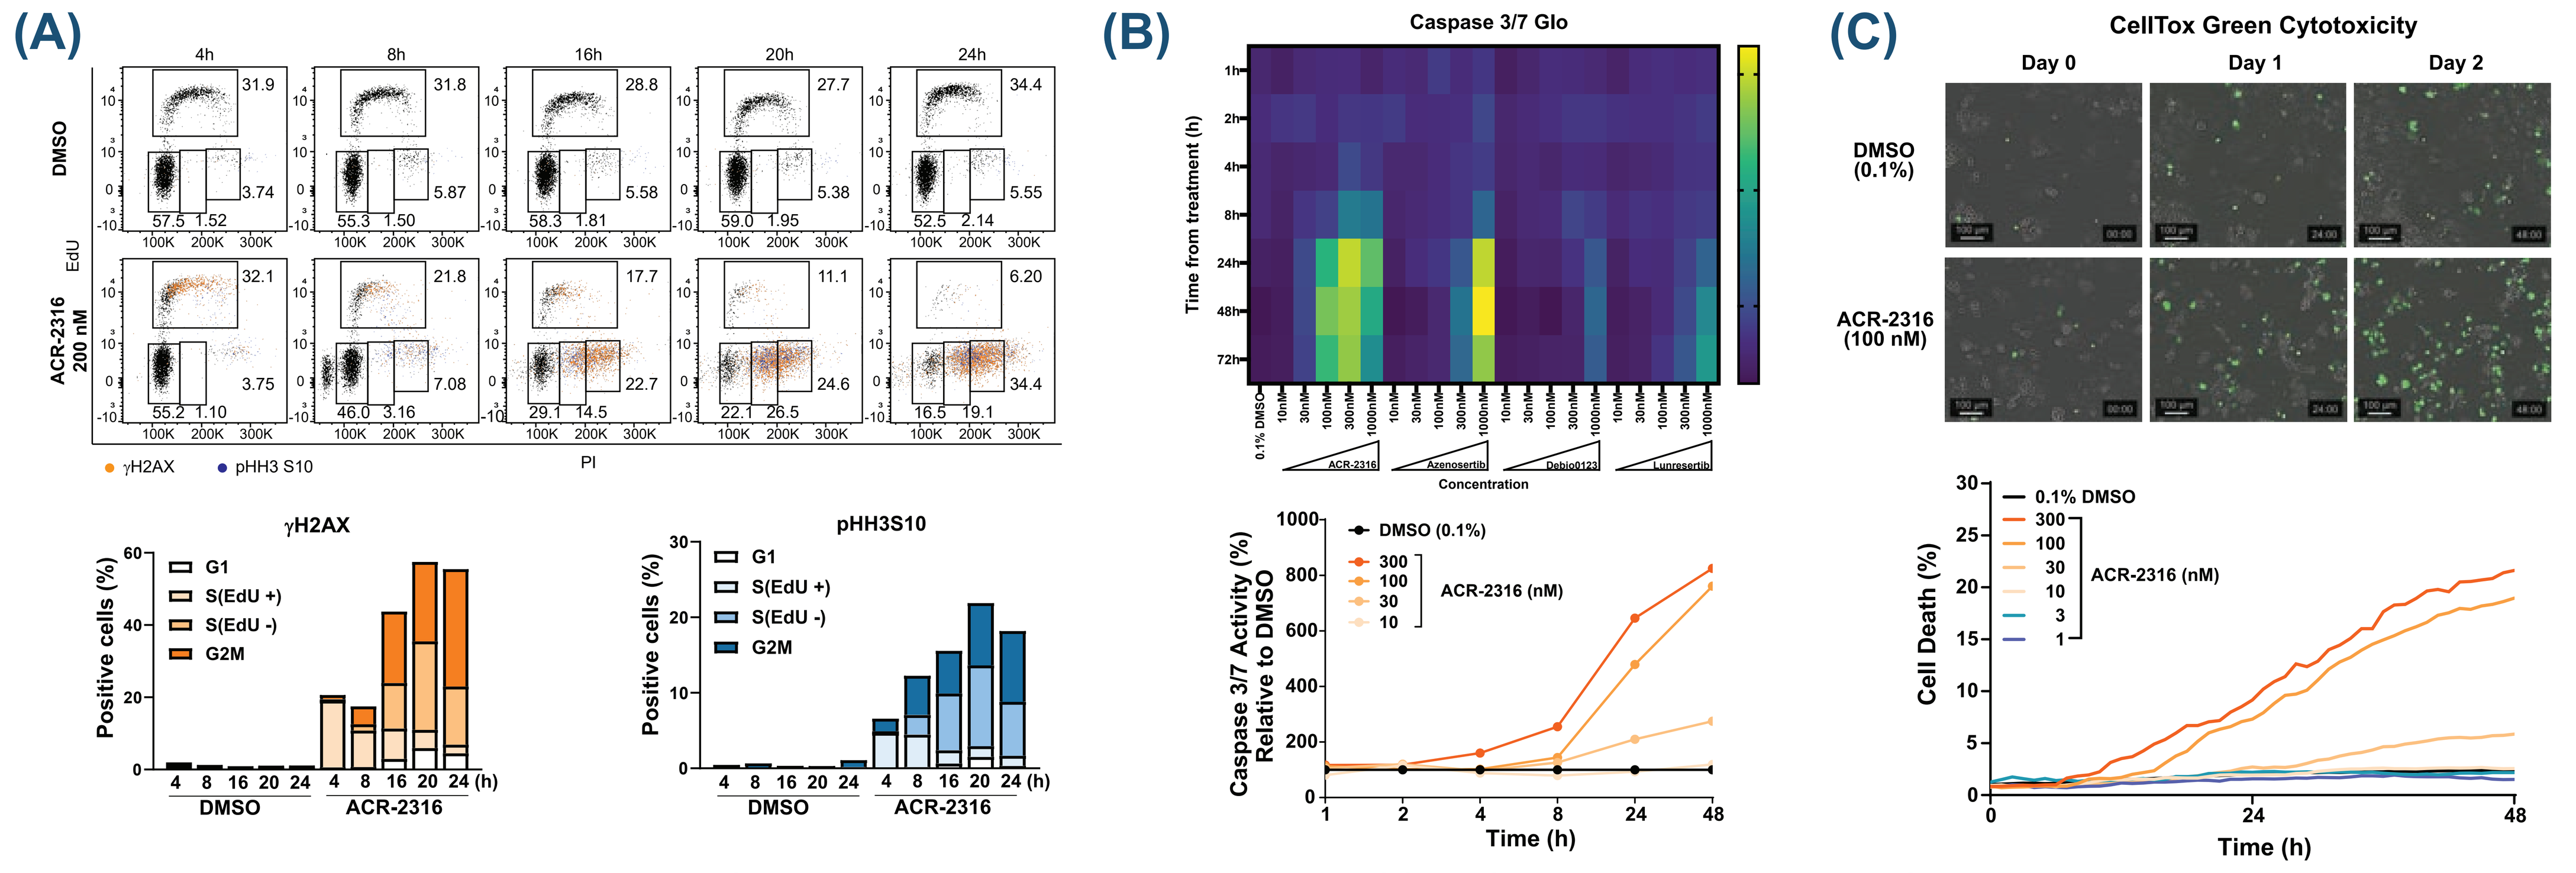
(A)
4h
8h
16h
20h
24h
DMSO
ACR-2316 200 nM
EdU
PI
γH2AX
pHH3 S10
Positive cells (%)
G1
S(EdU +)
S(EdU -)
G2M
(B)
Caspase 3/7 Glo
Time from treatment (h)
Concentration
ACR-2316
Azenosertib
Debio0123
Lunresertib
Caspase 3/7 Activity (%) Relative to DMSO
DMSO (0.1%)
ACR-2316 (nM)
Time (h)
(C)
CellTox Green Cytotoxicity
Day 0
Day 1
Day 2
DMSO (0.1%)
ACR-2316 (100 nM)
Cell Death (%)
0.1% DMSO
ACR-2316 (nM)
Time (h)

Balanced WEE1 and PKMYT1 inhibition by ACR-2316 drives unscheduled activation of CDK1/2, resulting in induction of DNA damage and premature mitotic cell death. (A) Ovarian cancer cells treated with 200 nM ACR-2316 display stalled DNA replication, induction of DNA damage (γH2AX) and premature mitotic signaling (pHH3S10) as assessed by flow cytometry at the indicated time points. (B) Induction of caspase 3-mediated apoptosis measured over time by Caspase-Glo assay reveals more potent induction by ACR-2316 in comparison to WEE1 and PKMYT1 inhibitors. (C) Live cell imaging using CellTox Green cytotoxicity assay shows cell death initiation 12h after ACR-2316 treatment and near-complete loss of viability by 48h.

Inhibition of CDK1 or CDK2 Reduces ACR-2316 Potency

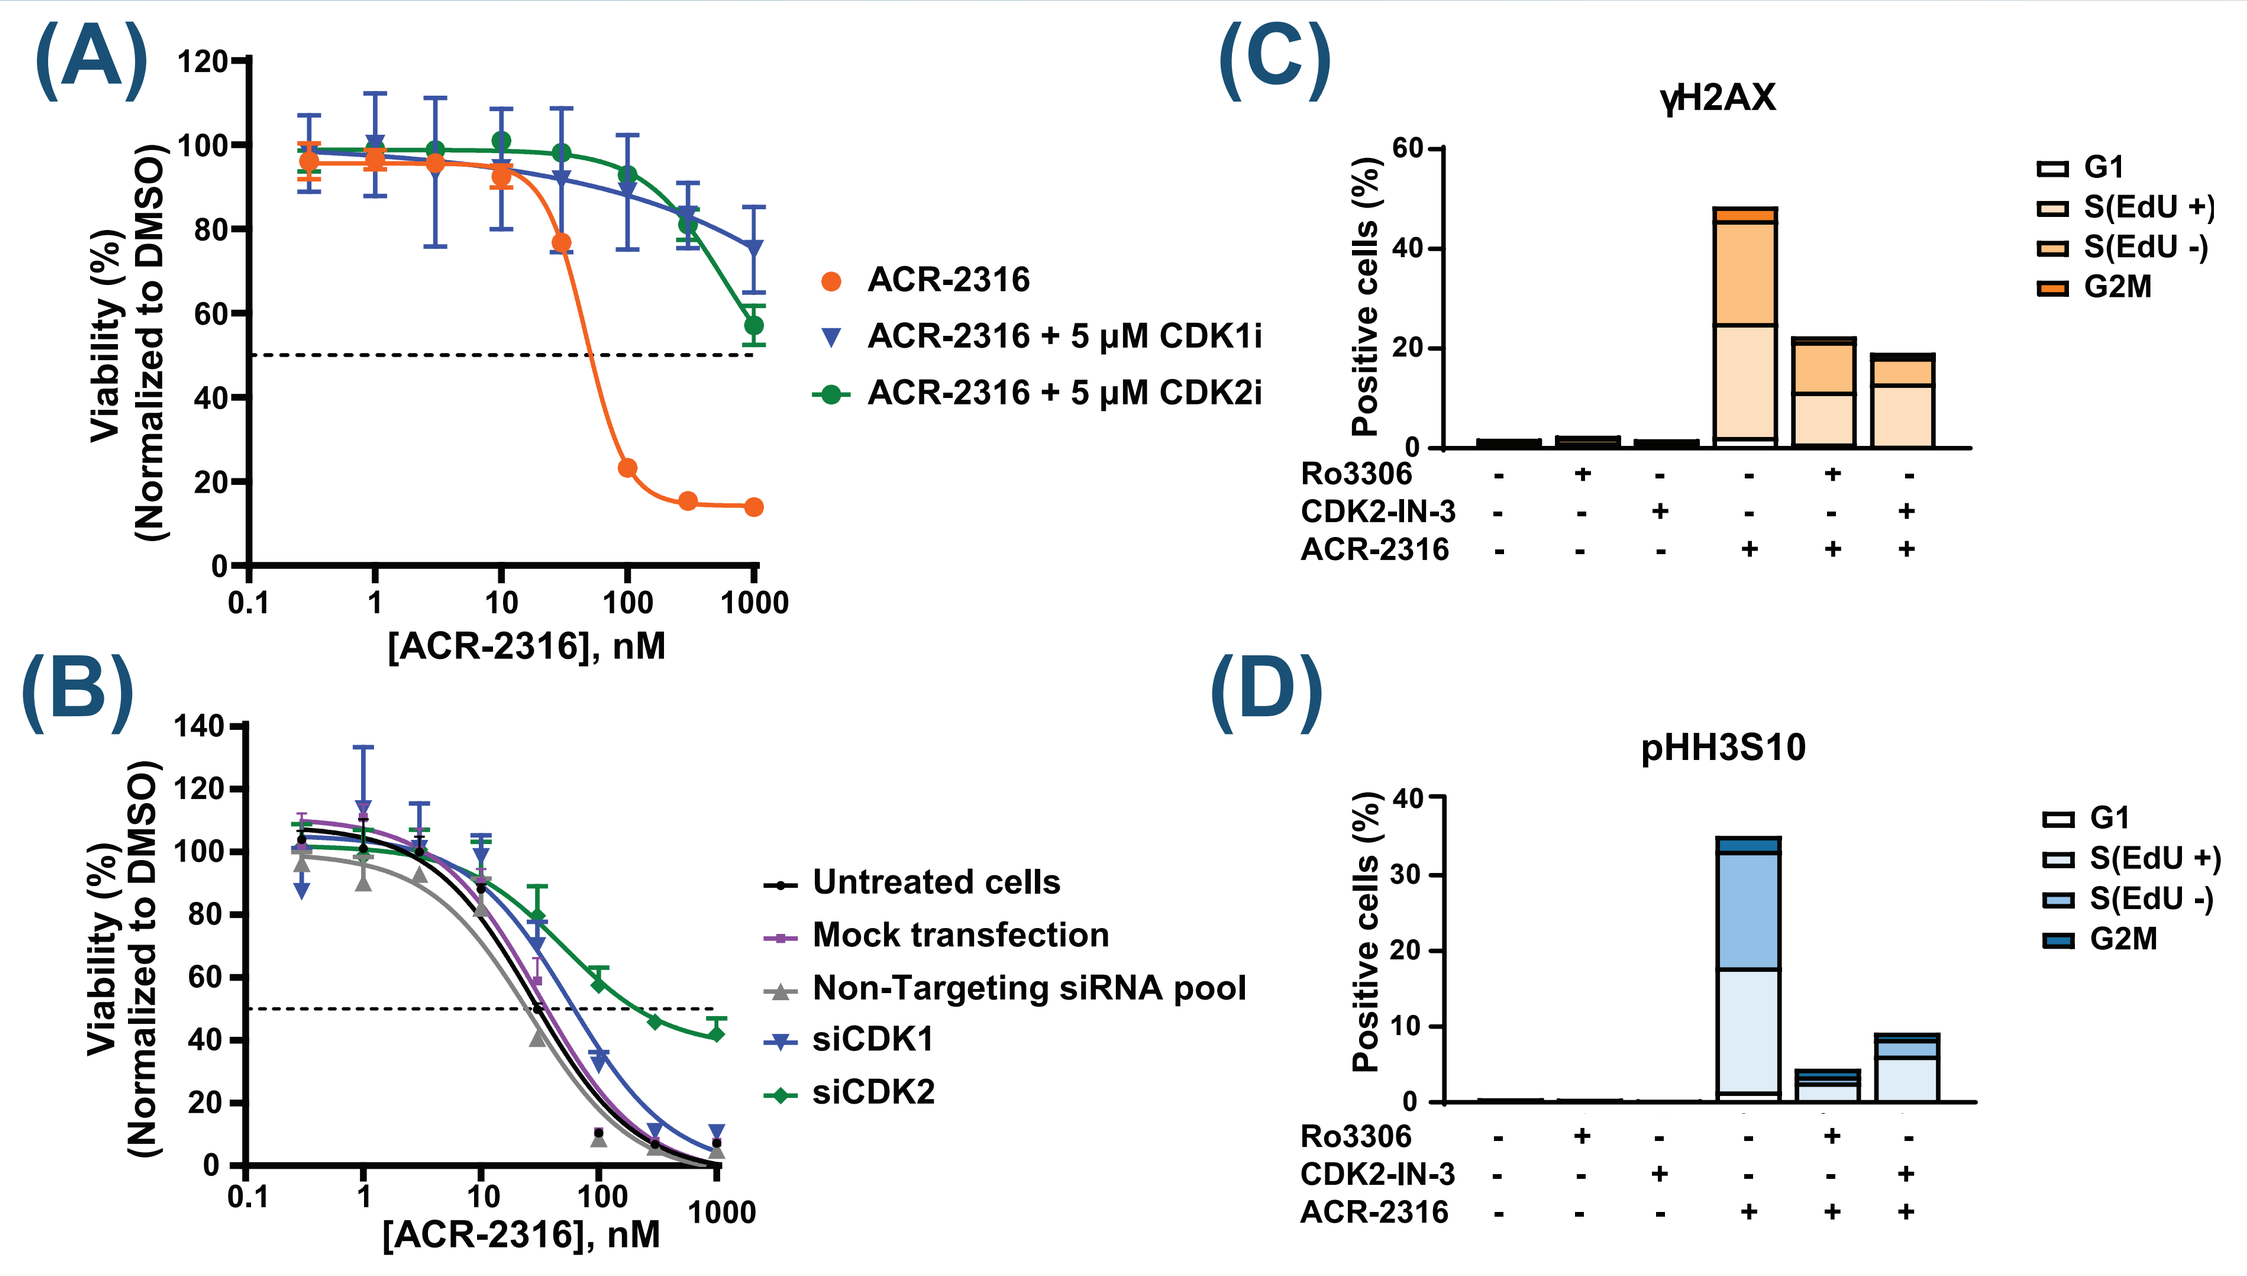
(A)
Viability (%) (Normalized to DMSO)
ACR-2316
ACR-2316 + 5 μM CDK1i
ACR-2316 + 5 μM CDK2i
[ACR-2316], nM
(B)
Untreated cells
Mock transfection
Non-Targeting siRNA pool
siCDK1
siCDK2
(C)
γH2AX
(D)
pHH3S10
Positive cells (%)
G1
S(EdU +)
S(EdU -)
G2M
Ro3306
CDK2-IN-3
ACR-2316

Inhibition of CDK1 or CDK2 limits ACR-2316 cell killing potency, confirming mechanism of action. Inhibition of CDK1 or CDK2 by pharmacological inhibition (A) or genetic knockdown (B) decrease ACR-2316 cell killing potency in CellTiterGlo viability assays. The phenotype observed in ovarian cancer cells treated for 4h with 200 nM ACR-2316, with accumulation of cells in S-phase, and induction of mitotic (C) and DNA damage (D) markers, was reverted by CDK1 or CDK2 inhibition, as quantified by flow cytometry.

ACR-2316 Activates PLK1 in Human Cancer Cells and Xenografts

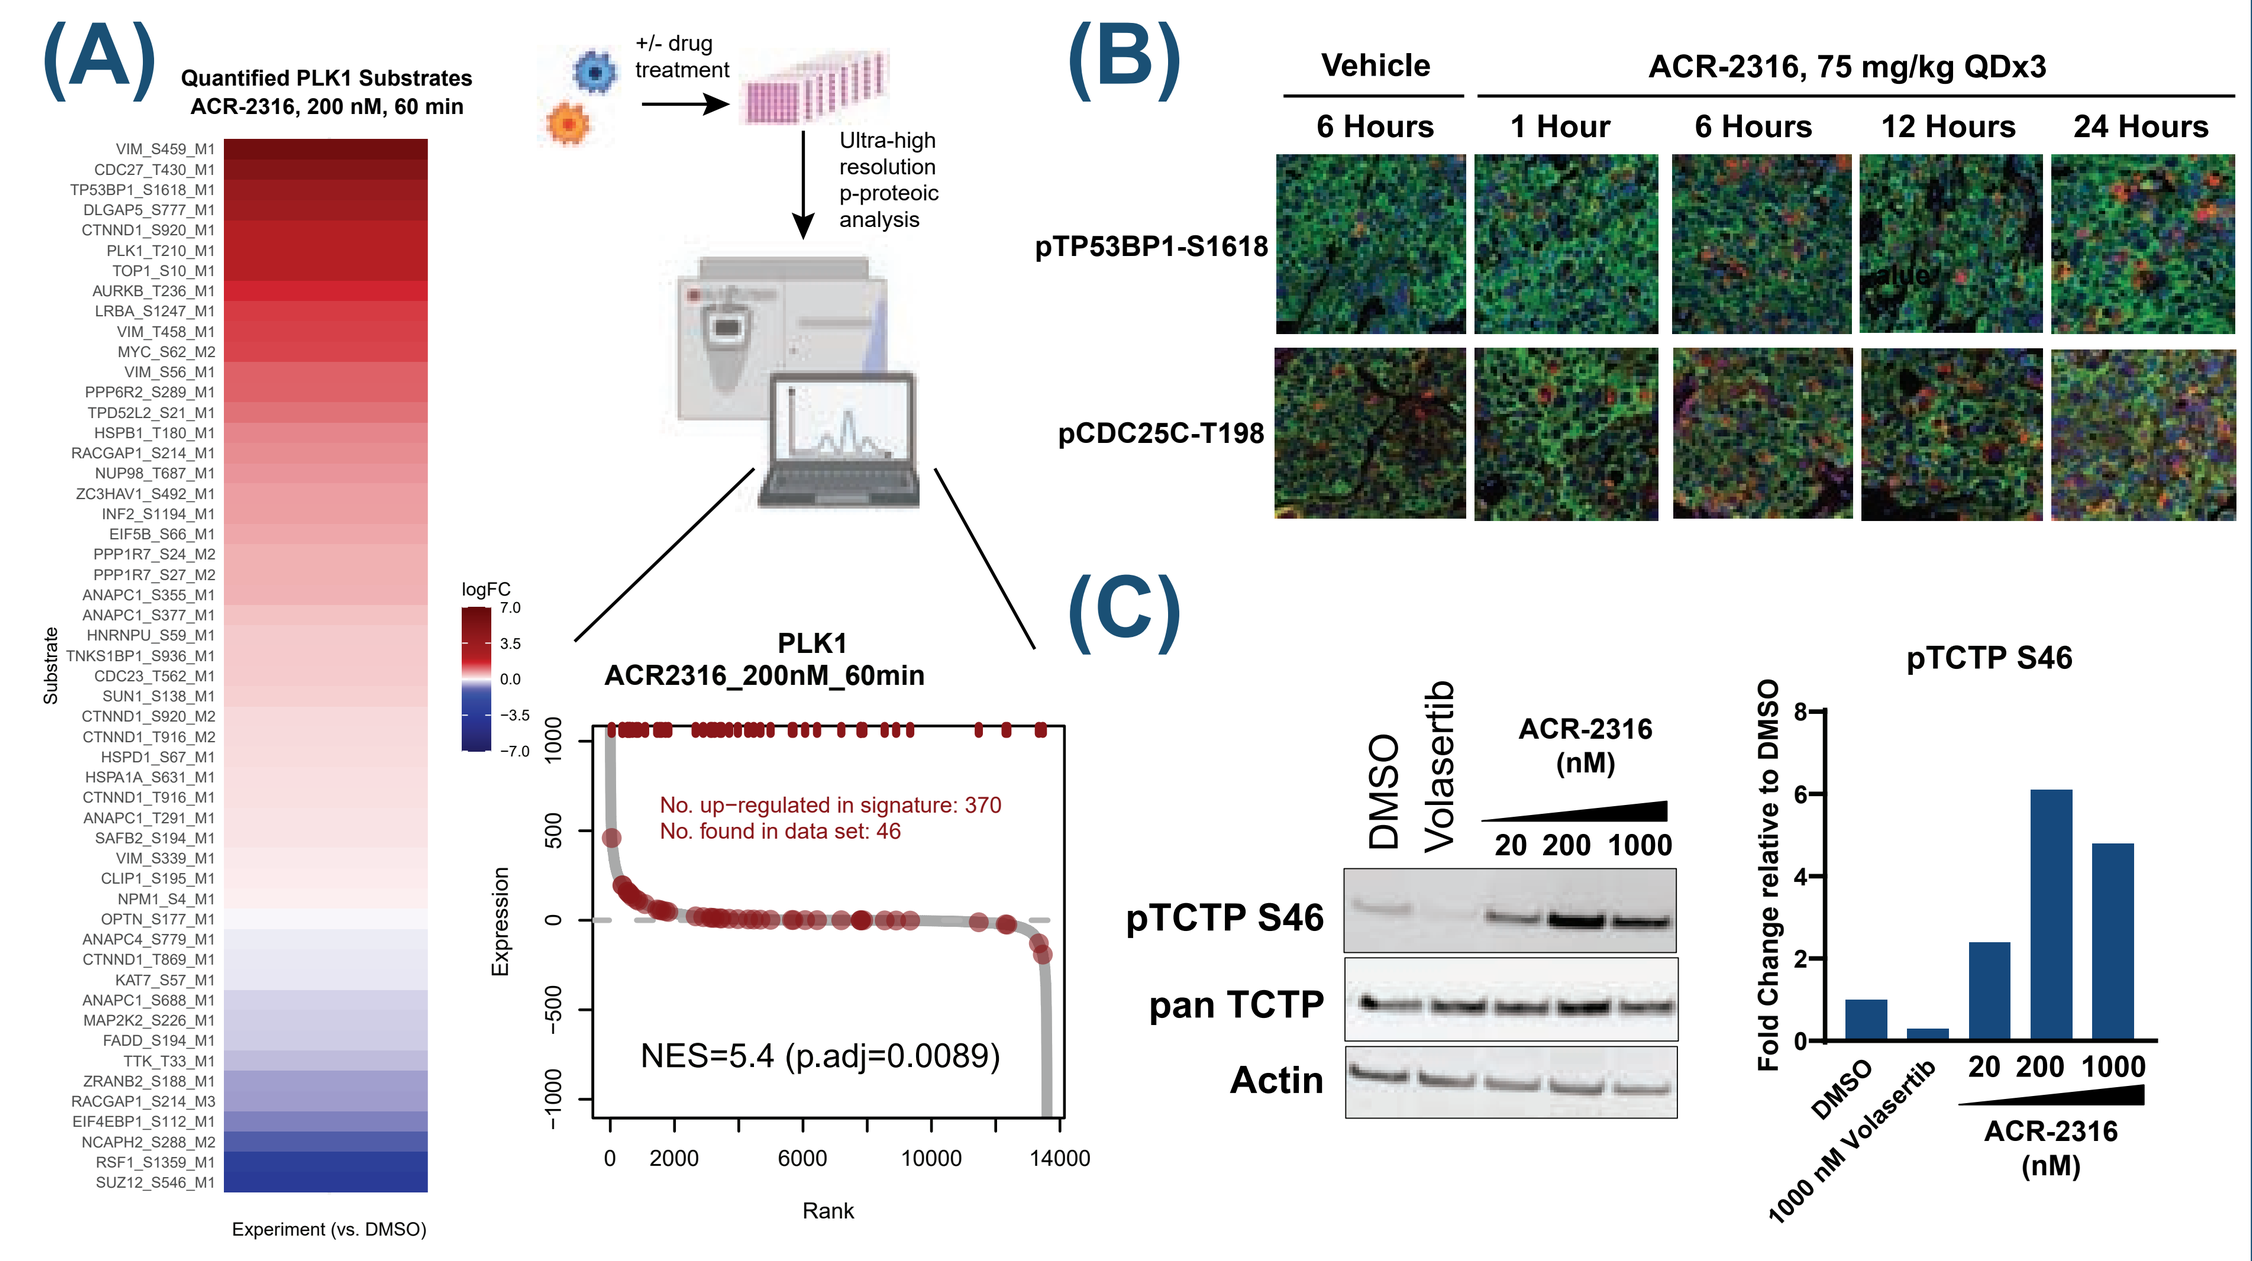
(A)
Quantified PLK1 Substrates ACR-2316, 200 nM, 60 min
+/- drug treatment
Ultra-high resolution p-proteoic analysis
PLK1
ACR2316_200nM_60min
No. up-regulated in signature: 370
No. found in data set: 46
NES=5.4 (p.adj=0.0089)
Rank
Expression
(B)
Vehicle
ACR-2316, 75 mg/kg QDx3
6 Hours
1 Hour
6 Hours
12 Hours
24 Hours
pTP53BP1-S1618
pCDC25C-T198
(C)
DMSO
Volasertib
ACR-2316 (nM)
20 200 1000
pTCTP S46
pan TCTP
Actin
Fold Change relative to DMSO
1000 nM Volasertib

ACR-2316 results in potent activation of PLK1. (A) Heatmap of ACR-2316 regulated PLK1 substrates (left) and PTM-SEA derived normalized enrichment score (right). (B) Immunofluorescence staining of ovarian cancer cell line-derived xenograft tumors from mice treated with ACR-2316 showing increased levels of PLK1 substrates pTP53BP1-S161 and pCDC25C-T198. (C) Western blot showing induction of PLK1 substrate pTCTP-S46 following 4h treatment with ACR-2316 in ovarian cancer cells.

ACR-2316 Induces Replication Stress, DNA Damage and Premature Mitosis in Xenograft Tumors

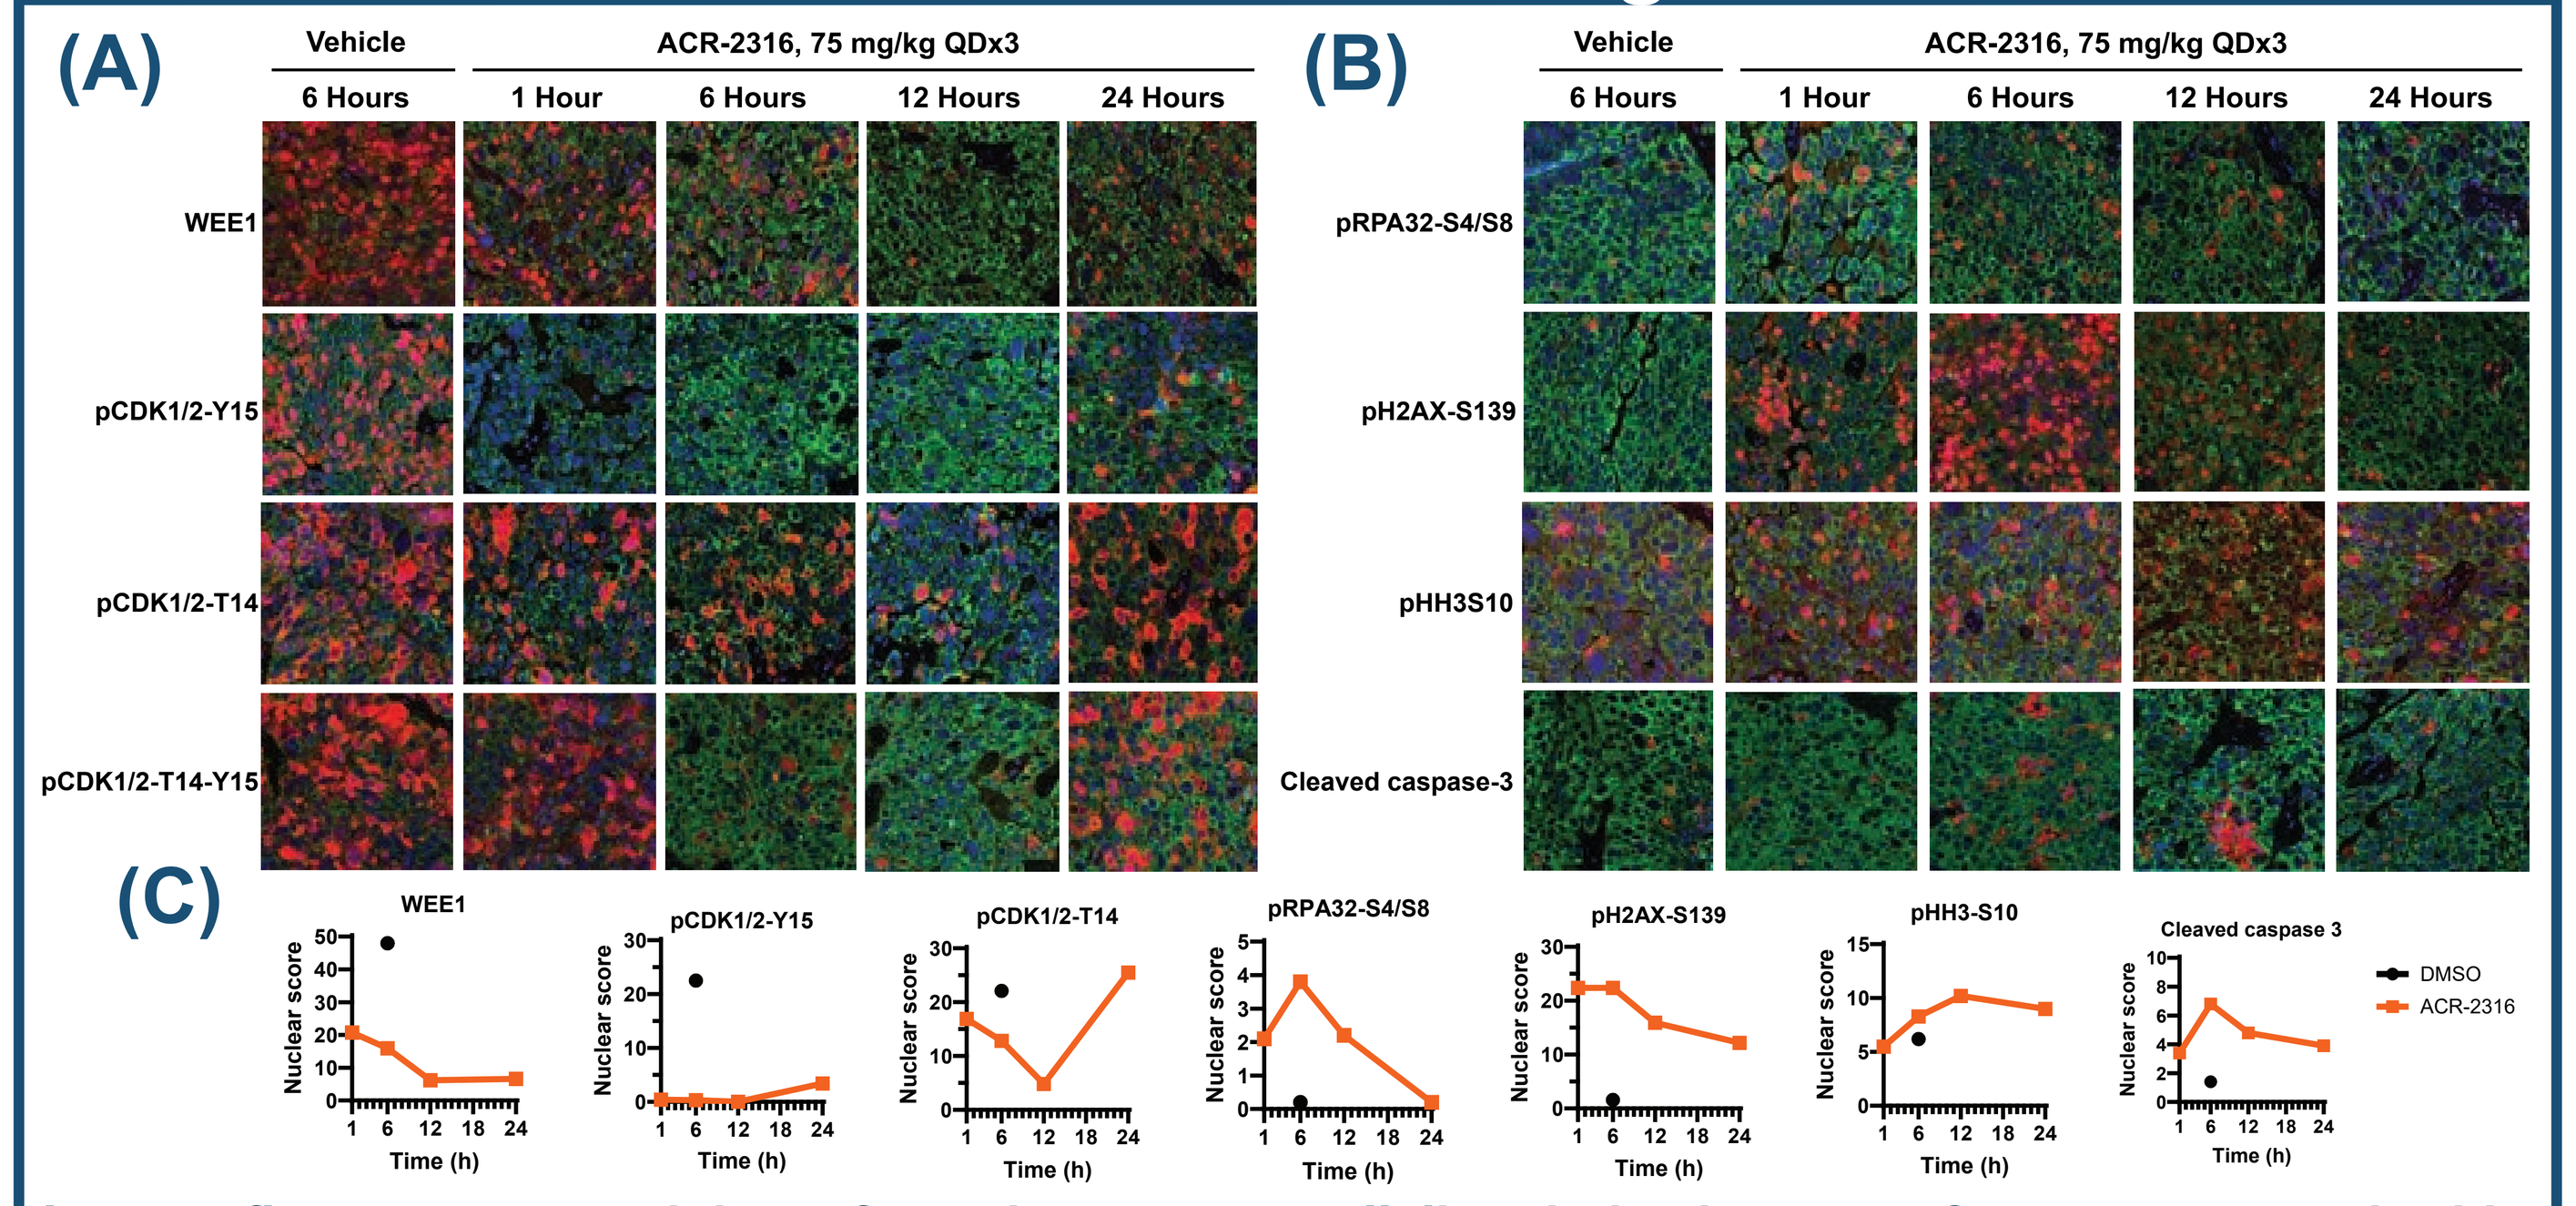
(A)
Vehicle
ACR-2316, 75 mg/kg QDx3
6 Hours
1 Hour
6 Hours
12 Hours
24 Hours
WEE1
pCDK1/2-Y15
pCDK1/2-T14
pCDK1/2-T14-Y15
(B)
pRPA32-S4/S8
pH2AX-S139
pHH3S10
Cleaved caspase-3
(C)
Nuclear score
Time (h)
DMSO
ACR-2316

Immunofluorescence staining of ovarian cancer cell line-derived xenograft tumors treated with ACR-2316 show increased replication stress, DNA damage and premature mitotic cell death. Immunofluorescence staining for (A) WEE1, pCDK1/2-Y15, pCDK1/2-T14, pCDK1/2-T14-Y15, and (B) markers of replication stress (pRPA32-S4/S8 and pH2AX-S139), mitosis (pHH3S10) and apoptosis (cleaved-caspase3). (C) Quantitative image analysis of nuclear score for each marker was evaluated in xenograft tumors 1-24h post-3-day treatment with ACR-2316 at 75 mg/kg.

ACR-2316 Demonstrates Dose-Dependent Efficacy and Superior Single Agent Activity

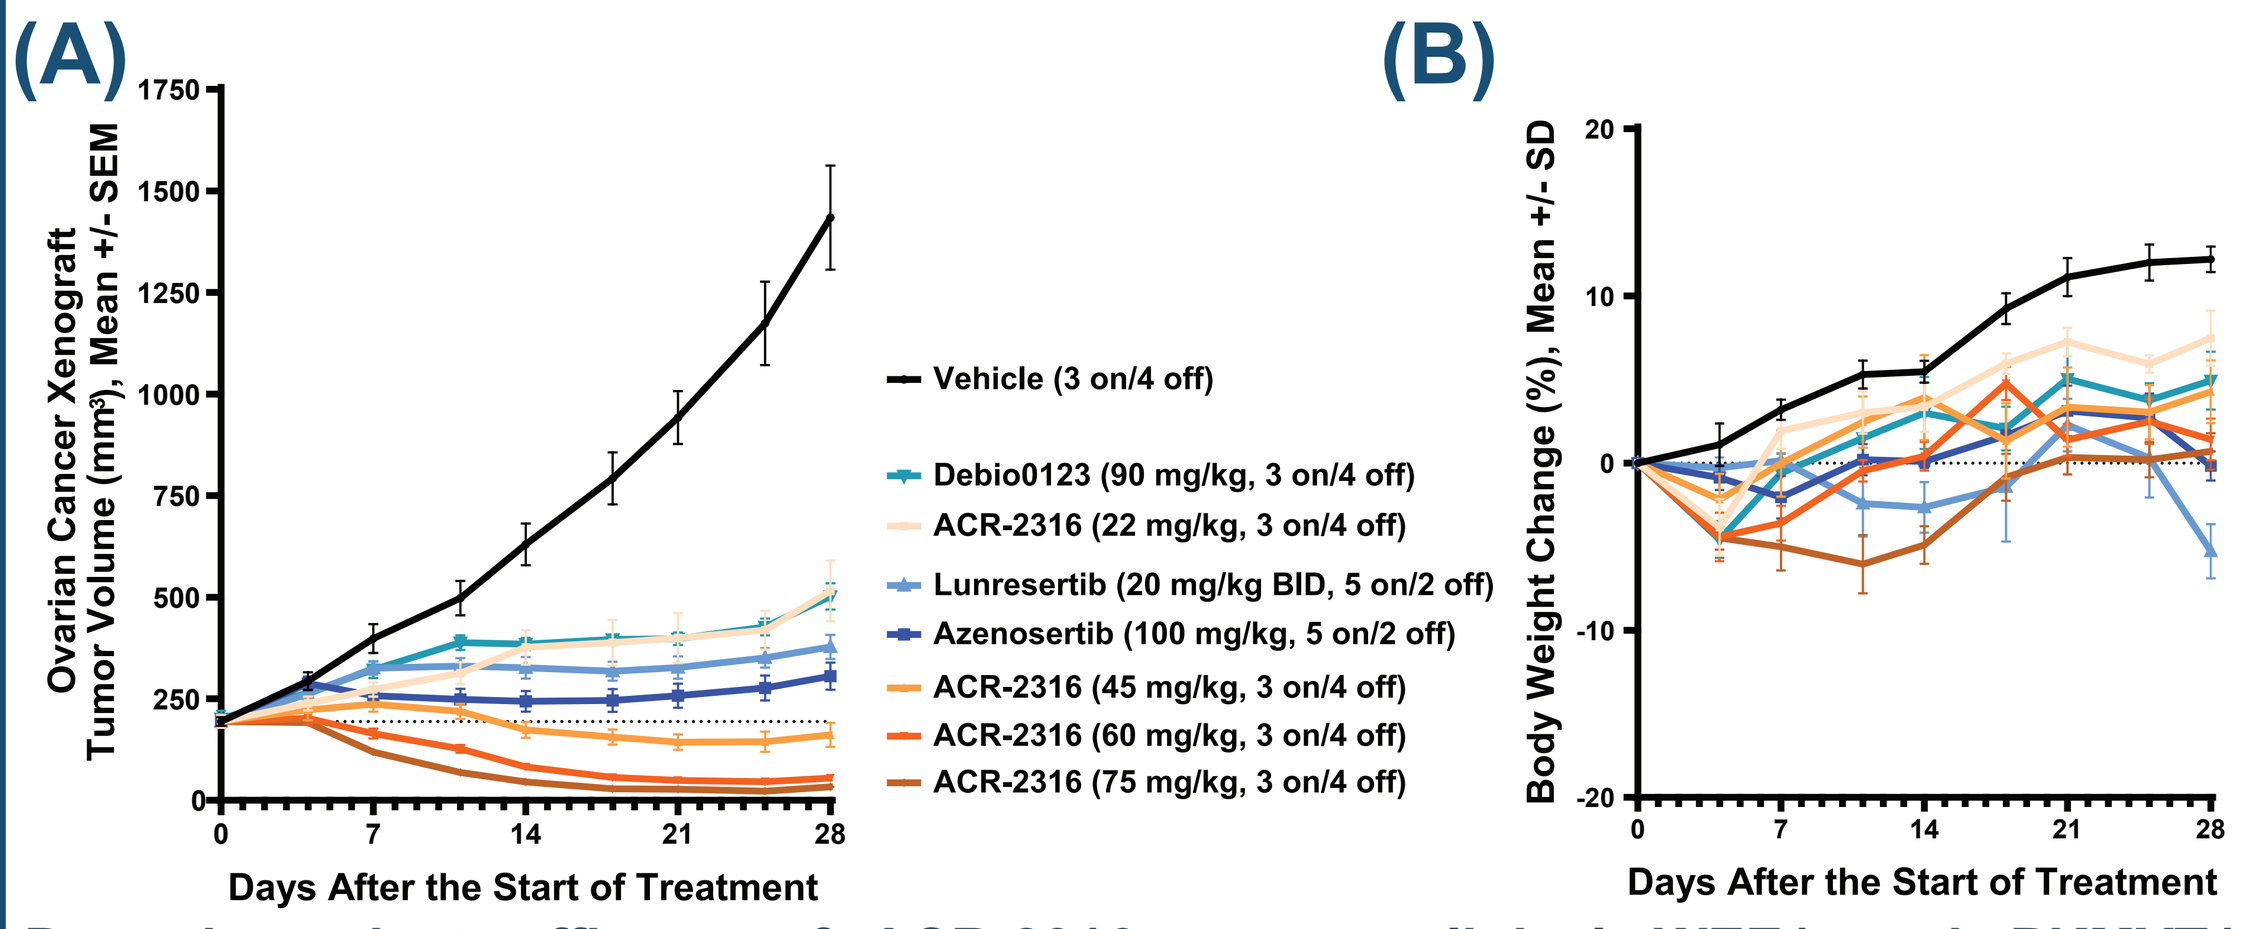
(A)
Ovarian Cancer Xenograft Tumor Volume (mm³), Mean +/- SEM
Vehicle (3 on/4 off)
Debio0123 (90 mg/kg, 3 on/4 off)
ACR-2316 (22 mg/kg, 3 on/4 off)
Lunresertib (20 mg/kg BID, 5 on/2 off)
Azenosertib (100 mg/kg, 5 on/2 off)
ACR-2316 (45 mg/kg, 3 on/4 off)
ACR-2316 (60 mg/kg, 3 on/4 off)
ACR-2316 (75 mg/kg, 3 on/4 off)
Days After the Start of Treatment
(B)
Body Weight Change (%), Mean +/- SD
Days After the Start of Treatment

Dose-dependent efficacy of ACR-2316 versus clinical WEE1 and PKMYT1 inhibitors. ACR-2316 demonstrates superior single agent efficacy in comparison to WEE1 inhibitors (azenosertib, Debio0123) and PKMYT1 inhibitor (lunresertib) in an ovarian cancer cell line-derived xenograft tumor model. Mice were dosed as indicated for 28 days. (A) Tumor volume represent Mean ± SEM. (B) Body weight change represent Mean ± SD.

ACR-2316 Promotes Durable Complete Response in Lung Cancer Xenograft Tumors

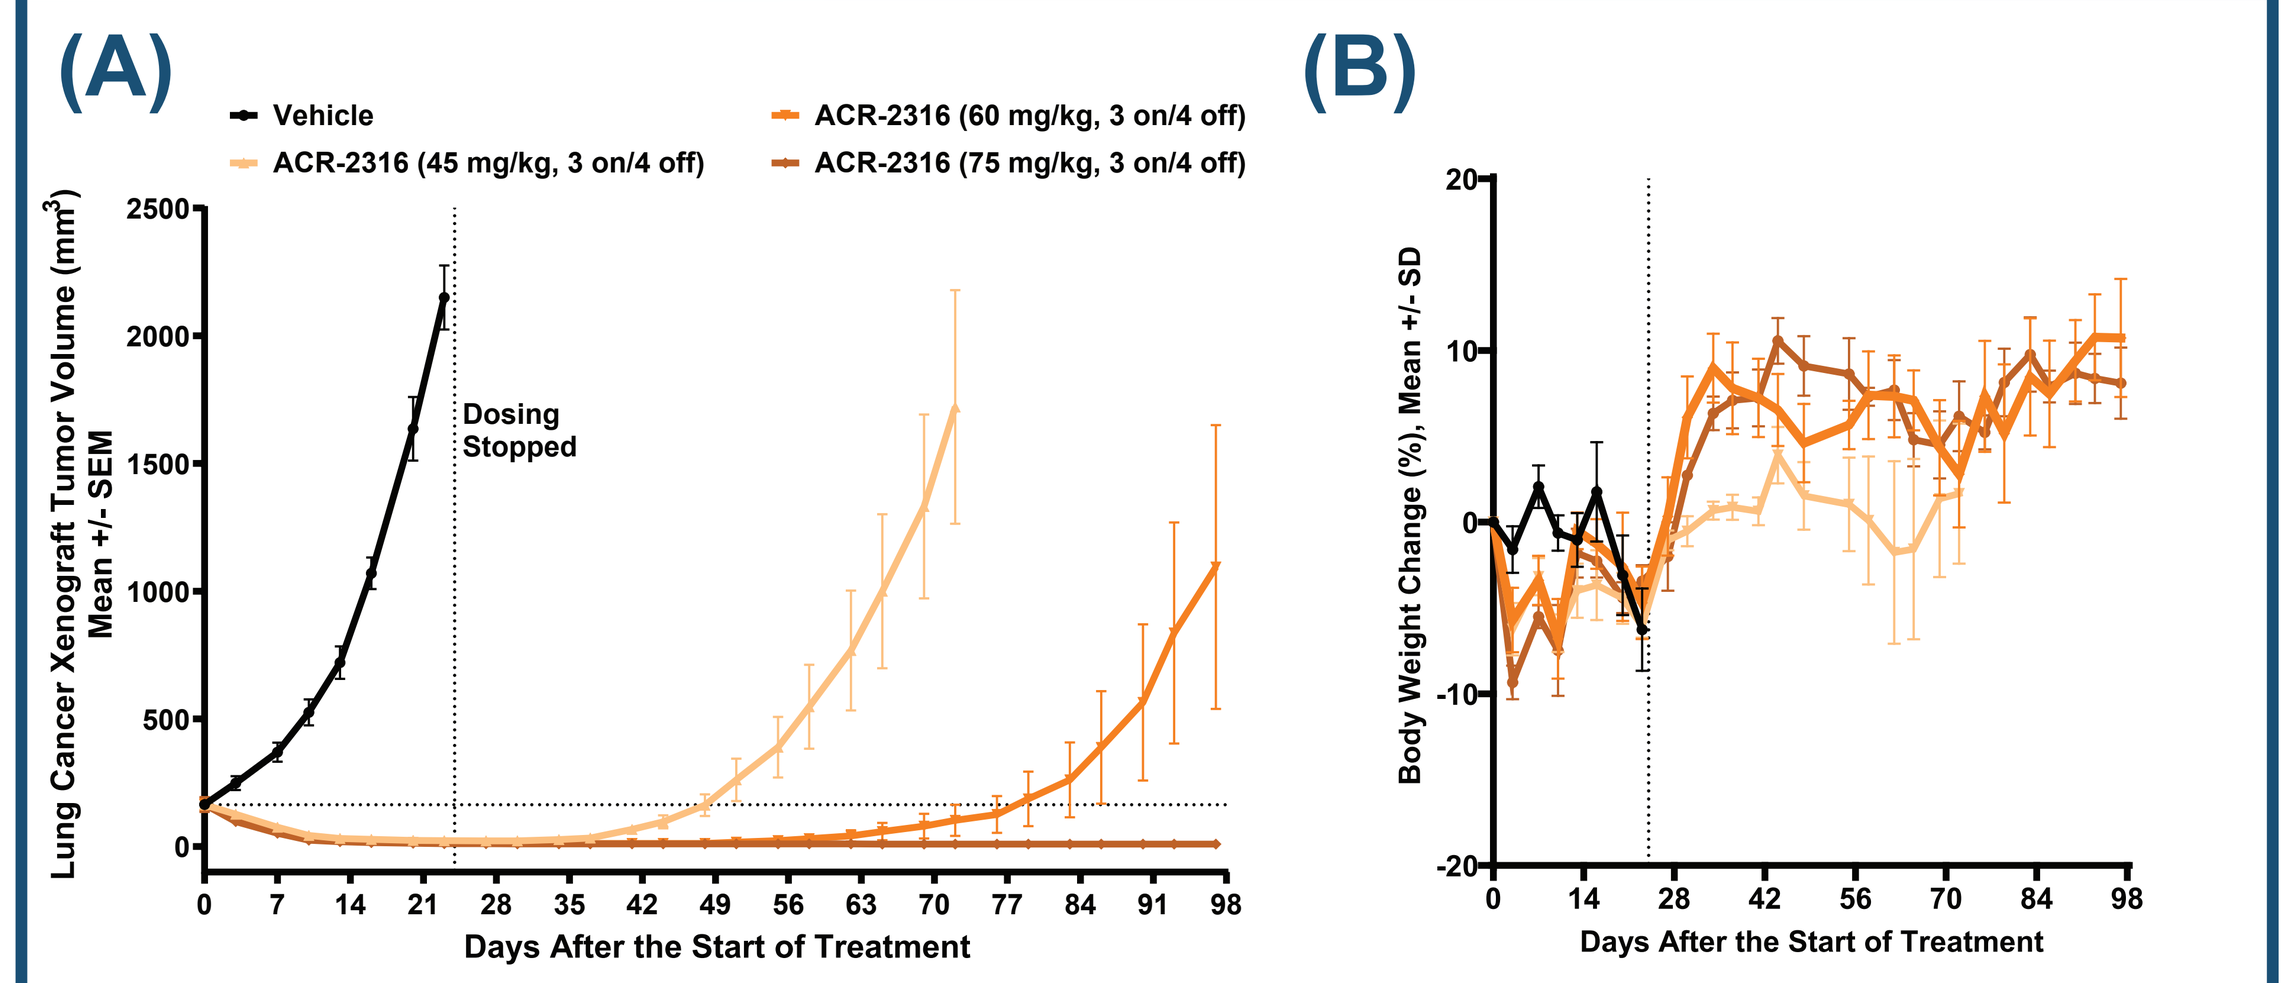
(A)
Vehicle
ACR-2316 (45 mg/kg, 3 on/4 off)
ACR-2316 (60 mg/kg, 3 on/4 off)
ACR-2316 (75 mg/kg, 3 on/4 off)
Lung Cancer Xenograft Tumor Volume (mm³) Mean +/- SEM
Dosing Stopped
Days After the Start of Treatment
(B)
Body Weight Change (%), Mean +/- SD
Days After the Start of Treatment

ACR-2316 leads to complete tumor regression in a lung cancer cell line-derived xenograft tumor model. ACR-2316 dosed orally for 3 weeks on a 3 on/4 off schedule leads to rapid, durable and complete tumor regression. (A) Tumor volume represent Mean ± SEM. (B) Body weight change represent Mean ± SD.

ACR-2316 Demonstrates Potent Anti-Tumor Activity in a Panel of 12 Ovarian PDX Tumor Models

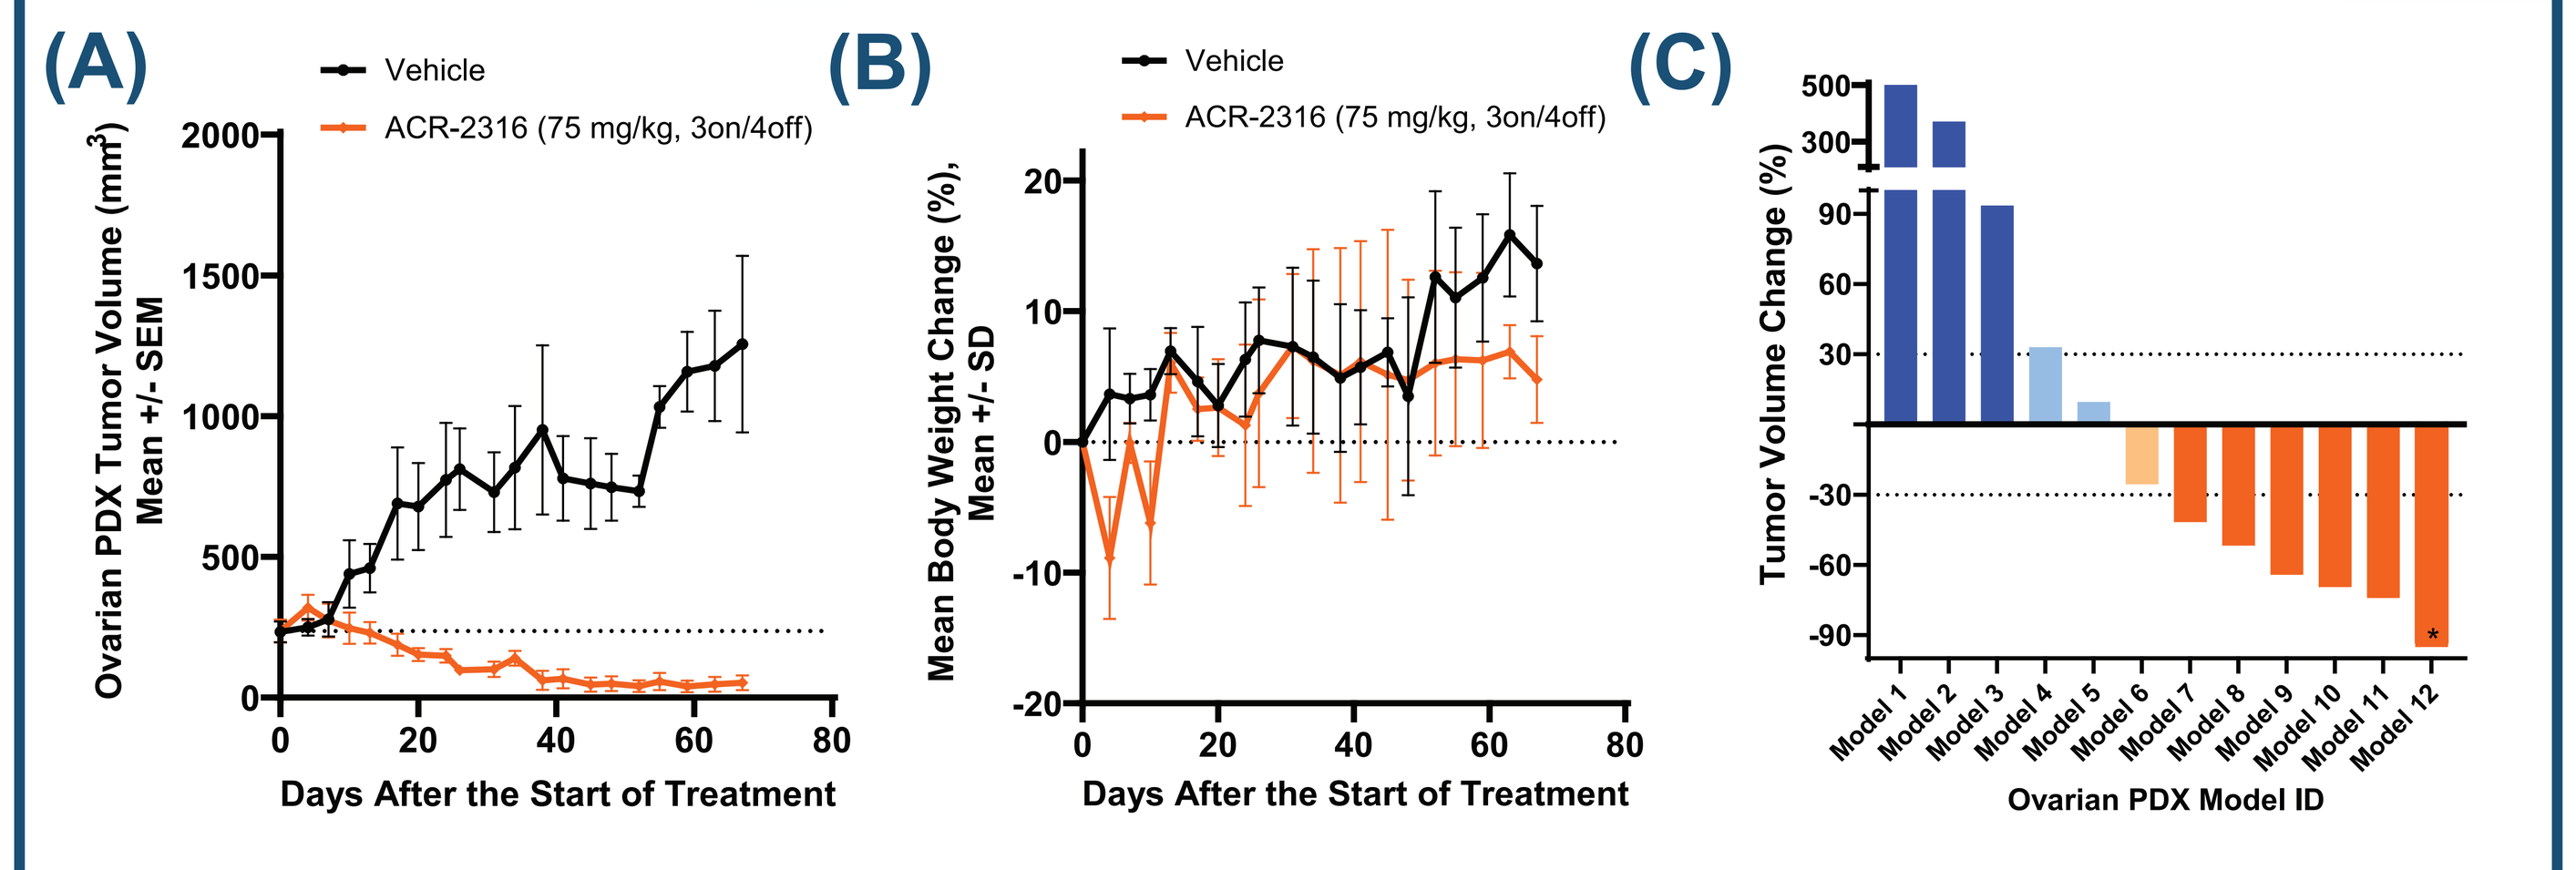
(A)
Vehicle
ACR-2316 (75 mg/kg, 3on/4off)
Ovarian PDX Tumor Volume (mm³) Mean +/- SEM
Days After the Start of Treatment
(B)
Mean Body Weight Change (%), Mean +/- SD
Days After the Start of Treatment
(C)
Tumor Volume Change (%)
Model 1
Model 2
Model 3
Model 4
Model 5
Model 6
Model 7
Model 8
Model 9
Model 10
Model 11
Model 12
Ovarian PDX Model ID

ACR-2316 is highly efficacious in the treatment of ovarian patient-derived xenograft tumor models. In an ovarian patient-derived xenograft tumor model, ACR-2316 dosed on a 3 on/4 off dosing schedule leads to rapid and potent tumor regression and is well-tolerated. (A)Tumor volume represent Mean ± SEM. (B) Body weight change represent Mean ± SD. (C) ACR-2316 demonstrates strong anti-tumor activity with frequent (5 on/2 off) and infrequent (*3 on/4 off) dosing in a panel of 12 ovarian PDX tumor models.

ACR-2316 Demonstrates Strong Efficacy Across a Broad Range of Intermittent Dosing Schedules

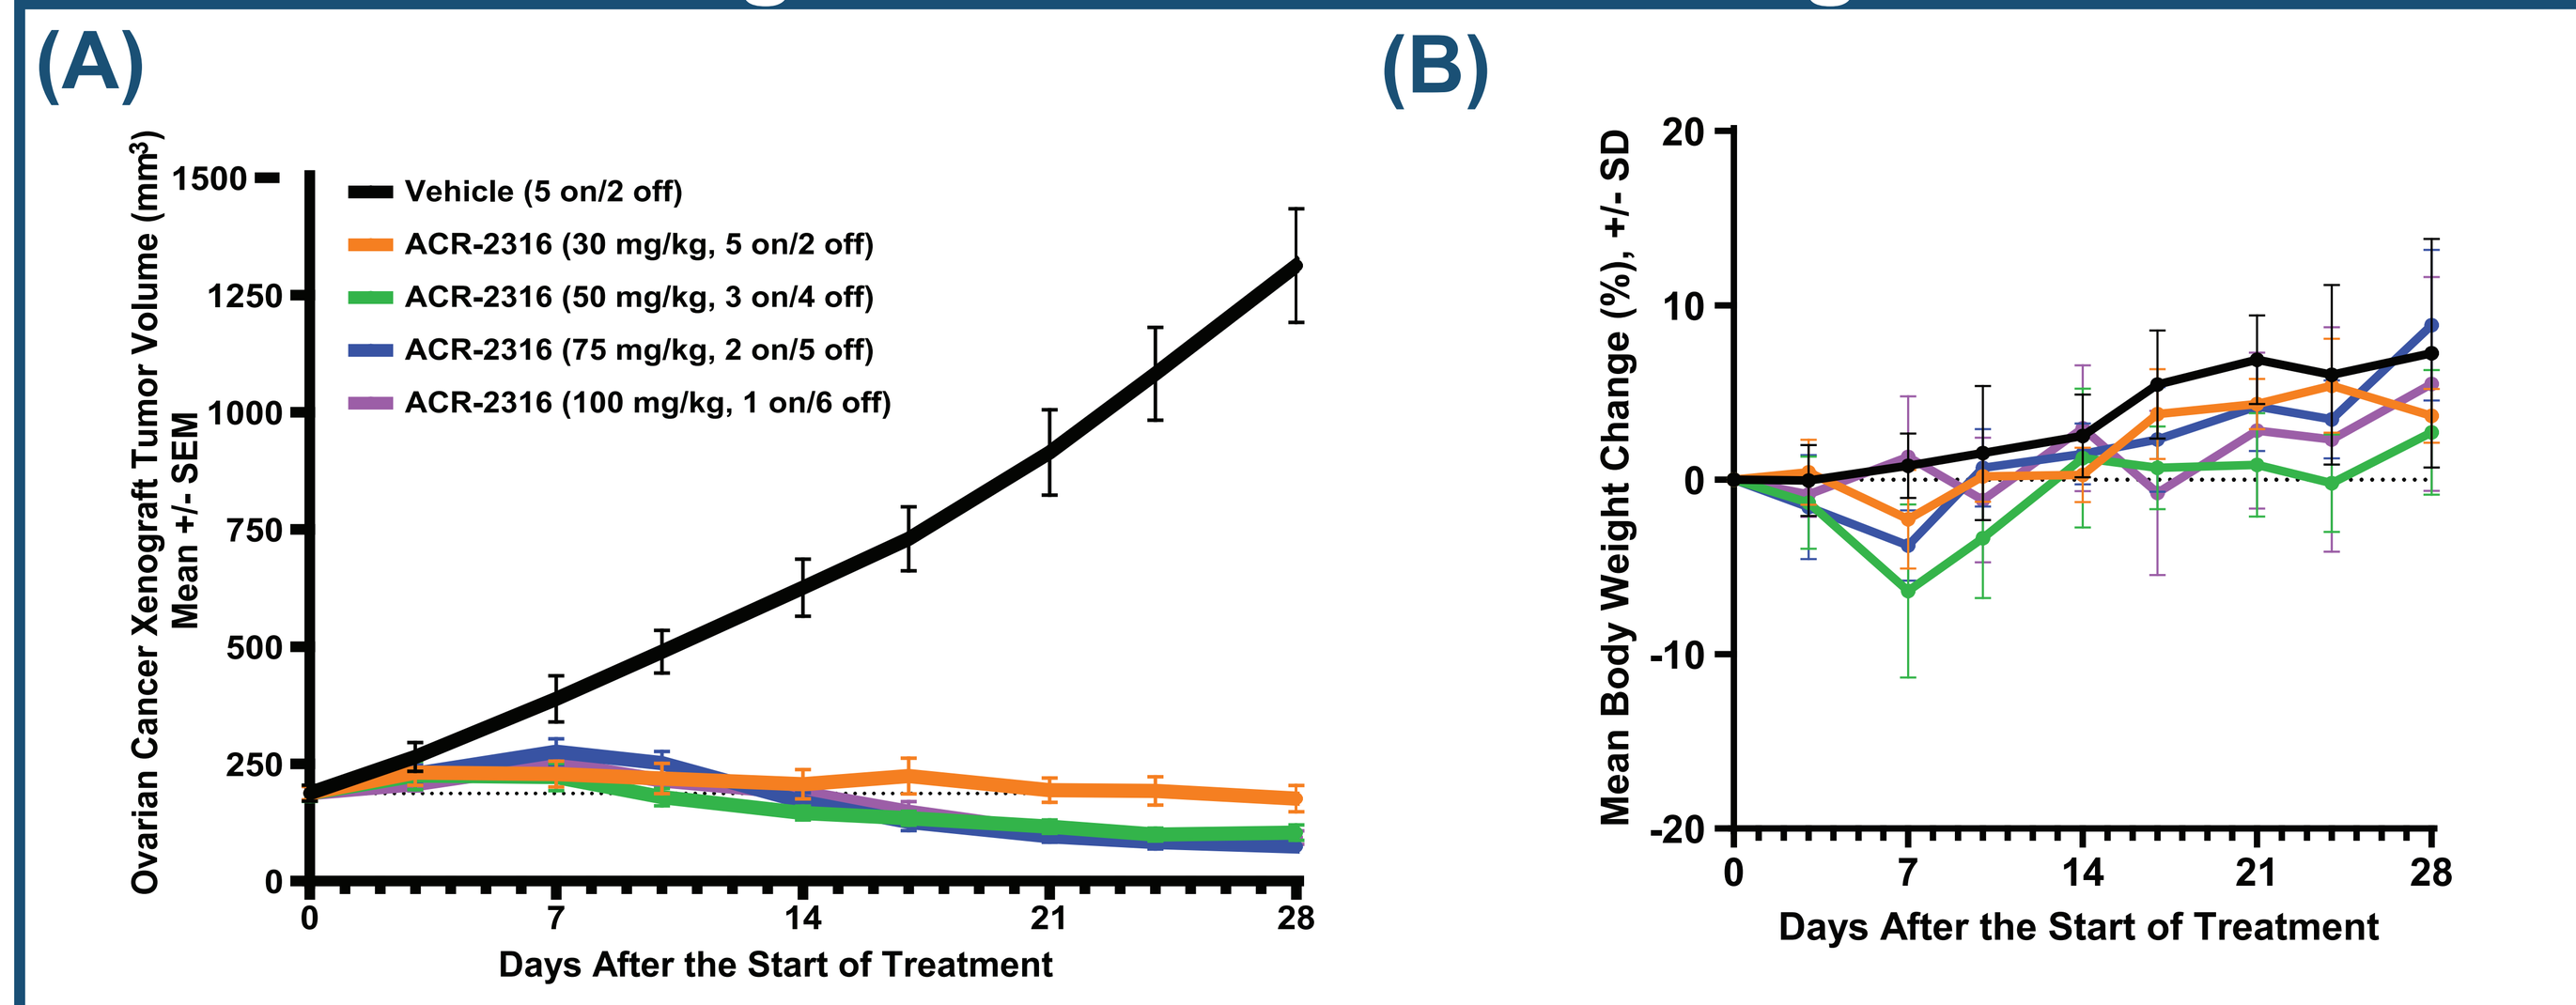
(A)
Ovarian Cancer Xenograft Tumor Volume (mm³) Mean +/- SEM
Vehicle (5 on/2 off)
ACR-2316 (30 mg/kg, 5 on/2 off)
ACR-2316 (50 mg/kg, 3 on/4 off)
ACR-2316 (75 mg/kg, 2 on/5 off)
ACR-2316 (100 mg/kg, 1 on/6 off)
Days After the Start of Treatment
(B)
Mean Body Weight Change (%), +/- SD
Days After the Start of Treatment

Anti-tumor activity of ACR-2316 with different dosing schedules was tested in a human ovarian tumor xenograft model. ACR-2316 was administered orally once, twice, for 3 days or 5 days per week. All doses tested demonstrated comparable efficacy and were well-tolerated. (A) Tumor volume represent Mean ± SEM. (B) Body weight change represent Mean ± SD.

Summary and Clinical Development

- **The unique capabilities of AP3 generates actionable insights with direct application for drug design optimization for monotherapy activity, and the identification of resistance mechanisms.**
- **ACR-2316 is a potent, selective dual inhibitor of WEE1 and PKMYT1, internally discovered, using biological structure-activity relationship uniquely enabled by AP3 to overcome the limitations of single-target inhibitors.**
- **ACR-2316 induces potent activation of CDK1, CDK2, and PLK1 resulting in stalled DNA replication, DNA damage, caspase 3-mediated apoptosis, and pro-apoptotic mitotic catastrophe.**
- **In CDX and PDX tumor models, ACR-2316 demonstrates dose-dependent efficacy and leads to complete tumor regression. Various ACR-2316 dosing schedules (QW, BIW, 3 days on/4 days off, 5 days on/2 days off) also regress tumors, providing clinical flexibility.**
- **ACR-2316 is well-tolerated in rodents, demonstrating only mechanism-based, transient, and reversible myelosuppression across a range of intermittent dosing regimens, resulting in potent anti-tumor activity and a favorable therapeutic index for human dosing.**
- **Patient dosing has begun in the ongoing Phase 1 monotherapy clinical trial designed to assess the safety and tolerability of ACR-2316.**

ACRIVON THERAPEUTICS